Safety and Efficacy of BRAF Inhibitor Plixorafenib (FORE8394; PLX8394) in Children and Adults With Recurrent, BRAF-altered Primary Central Nervous System Tumors (PCNST)

Macarena de la Fuente[1], Nicholas A Butowski[2], Jennie Taylor[2], Rona Yaeger[3], Frank Y-C Tsai[4], Filip Janku[5], Carl E Allen[6], Natraj Ammakkanavar[7], Glenn Michelson[8], Ping Jiang[9], Emil Samara[10], Michael Paz[9], Alexia Tussay-Lindenberg[9], Kongming Wang[9], Stacie Peacock Shepherd[9], Irina Kline[9], Eric Sherman[3], Jordi Rodon[5]

[1]Sylvester Comprehensive Cancer Center, University of Miami, Miami, FL, USA; [2]University of California, San Francisco, San Francisco, CA, USA; [3]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [4]HonorHealth Research Institute, Scottsdale, AZ, USA; [5]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [6]Baylor College of Medicine, Texas Children's Hospital, Houston, TX, USA; [7]Community Health Network, Cancer Center, Indianapolis, IN, USA; [8]Audentes Consulting, Fairfax, CA, USA; [9]Fore Biotherapeutics, Philadelphia, PA, USA; [10]PharmaPolaris International Inc, San Marino, CA, USA

BACKGROUND

Plixorafenib

- A potent, orally available BRAFi that selectively targets V600 and non-V600 alterations through disruption of BRAF monomers and dimers.[1]
- Evades paradoxical MAPK pathway activation, eliminating the need for coadministration with a MEK inhibitor.[2]
- Designed to be more selective and tolerable than early-generation BRAFis.[1,3,4]

High Unmet Need for CNS Tumors

- In children, the 5-year relative survival rate for primary CNS tumors is 68.9%.[5]
- Primary CNS cancers represent the most common cause of death in children.[5]

METHODS

Study Description

- Phase 1/2a, open-label, single-arm, multicenter study (NCT02428712) to assess the safety, PK, and preliminary efficacy of oral plixorafenib 900–3600 mg/day with or without cobicistat[a] (PK enhancer) in children and adults with BRAF-altered solid tumors.
 - Phase 1 in adults and phase 1 in children 4–17 years (n=4)[b] who received BSA-adjusted dosing, both with 3+3 design
 - Phase 2a: dose optimization and extension in patients aged ≥10 years

Overall patient population[c] (N=113)
- 64 (57%) had V600+ disease
- 34 (30%) had received ≥1 prior MAPK-targeted regimen

Prespecified efficacy subgroups[d]
- BRAF V600+: N=43[e]
- BRAF fusions: N=14
- PCNST BRAF V600+, MAPKi naïve: N=9
- Phase 1 pediatric: N=4

Key eligibility criteria
- Histologically confirmed, advanced, unresectable solid tumors
- Measurable disease by RECIST or RANO criteria
- Intolerant to standard therapy or no standard therapy available
- No symptomatic brain metastases

PCNST[f] (safety subgroup, n=22)
- 15 (68%) had a V600+ BRAF mutation
- 13/15 were V600+ with ≥1 postbaseline assessment

Pediatric population (n=5)[g]
- 2 (40%) had a V600+ BRAF mutation (1 LCH-ND, 1 LGG)
- 2 (40%) received ≥1 prior MAPK therapy (1 LCH-ND, 1 LGG)

[a]Cobicistat was used in adult patients as a PK enhancer with no anticancer effect.
[b]One pediatric patient was enrolled into phase 2.
[c]All patients who received ≥1 dose of plixorafenib.
[d]mITT population: patients who received ≥1 dose of plixorafenib, with BRAF Class I or II alterations, with any tumor evaluation data following the first dose of plixorafenib.
[e]Excludes patients with CRC due to known intrinsic resistance through the EGFR pathway.
[f]For patients with PCNST, tumor assessments were performed using RECIST 1.1 or RANO HGG or LGG criteria based on the protocol amendment under which the patient was enrolled.
[g]Phase 1 in pediatric patients was completed (not due to safety or efficacy concerns) and pediatric patients were eligible for phase 2.

RESULTS

Baseline Characteristics

Demographics		Overall (N=113)	PCNST (n=22)
Age, y	Median (min, max)	57.0 (4, 86)	41.5 (4, 80)
Sex, n (%)	Male	59 (52.2)	9 (40.9)
Race, n (%)	White	101 (89.4)	18 (81.8)
	Black/African American	5 (4.4)	2 (9.1)
	Asian	3 (2.7)	1 (4.5)
	Missing	4 (3.5)	1 (4.5)
Ethnicity, n (%)	Hispanic or Latino	12 (10.6)	5 (22.7)
ECOG performance status, n (%)	0	43 (38.1)	11 (50.0)
	1	65 (57.5)	9 (40.9)
	≥2	5 (4.4)	2 (9.1)
Prior lines of therapy, n (%)	0	15 (13.3)	6 (27.3)
	1	28 (24.8)	7 (31.8)
	2	16 (14.2)	0
	3	17 (15.0)	5 (22.7)
	≥4	37 (32.7)	4 (18.2)
Prior MAPK-targeted therapies, n (%)	Any MAPK-targeted therapy	34 (30.1)	5 (22.7)
	>1 prior MAPKi[a]	13 (38.2)	1 (20.0)
	BRAFi[a]	24 (70.6)	3 (60.0)
BRAF mutation, n (%)	V600 (Class I)	64 (56.6)	15 (68.2)
	V600E	60 (53.1)	15 (68.2)
	Other[b]	4 (3.5)	0
	Class II	36 (31.9)	6 (27.3)
	Fusion	17 (15.0)	5 (22.7)
	Nonfusions	19 (16.8)	1 (4.5)
	Class III	1 (0.9)	0

[a]Percentage is based on the total number of any MAPK-targeted therapy.
[b]Includes V600K and V600R variants.

- **Tumor Type:** 99.1% solid tumors, including 8.8% LGG/GNT and 10.6% HGG; 1 (0.9%) patient with LCH-ND

Disposition

- Overall population (N=113)
 - 89.2 patient-years of cumulative plixorafenib exposure
 - 106 (93.8%) patients discontinued treatment due to RECIST/RANO-defined PD or clinical disease progression (n=87), patient withdrawal (n=9), AE (n=5; 1 discontinued due to treatment-related AE [grade 3 bilirubin; dose: 1800 mg BID with PK enhancer]), other reason (n=3), physician decision (n=1), death (n=1)
- PCNST population (n=22)
 - 16.4 patient-years of cumulative plixorafenib exposure
 - 20 (90.9%) patients discontinued treatment due to RECIST/RANO-defined PD or clinical disease progression (n=17), patient withdrawal (n=1), AE (n=1), other reason (n=1)

Conflicts of Interest

MdlF: Advisory board: Fore Biotherapeutics (Fore Bio), Servier, AnHeart, Rigel. Research support: Caris. Board of Directors: Neuro-Oncology. Receipt of equipment, materials, drugs, medical writing, gifts or other services: Fore Bio. **JT**: Grants: BMS, Servier. Consultant: Servier, University of Colorado, Mt Sinai Health System. Editor: UpToDate. **RY**: Grants: Boehringer Ingelheim, Boundless Bio, Daiichi Sankyo, Pfizer. Personal fees: Revolution, Loxo@Lilly, Zai Lab. Grants and personal fees: Mirati. **FT**: Honoraria: Aptitude Health. Stock/stock options: Salarius, Sphinx Health Solutions Corp. **FJ**: Employee and stock/stock options: Monte Rosa. Advisor: Fore Bio. **CEA**: Grants/contracts: NCI/NIH, St. Baldrick's Foundation, The Leukemia & Lymphoma Society. Consultant: Sobi, Electra, OPNA. **NA**: Honoraria: BMS. Meeting support: Loxo Oncology. Stock/stock options: Iovance. **GM**: Consultant: Fore Bio. **PJ**, **MP**, **ATL**, **KW**, **IK**: Employee: Fore Bio. **ES1**: Consultant: Fore Bio. **SPS**: Employee: Fore Bio. Stock: Fore Bio, Abbott, AbbVie, BridgeBio. **ES2**: Grants: Fore Bio, Eli Lilly, Regeneron, Pure Tech Health. Personal fees: AffyImmune, Eli Lilly. **JR**: Grants/contracts: Blueprint Medicines, Merck Sharp & Dohme, 280 Bio. Consultant: Cancer Core Europe, Hummingbird, Yingli, Merus, Aadi, Fore Bio, Amgen, Monte Rosa, Debio, Incyte, BridgeBio, Vall d'Hebron Institute of Oncology/Cancer Core Europe. Meeting support: Loxo Oncology. Data safety monitoring/advisory board: Ellipses, Molecular Partners, iOnctura, Sardona, Mekanistic, Amgen, Merus, Monte Rosa, Aadi, BridgeBio. Other: Cancer Core Europe, Symphogen, BioAtla, Pfizer, Kelun-Biotech, GSK, Taiho, Roche, Hummingbird, Yingli, Bicycle, Merus, Aadi, Fore Bio, Loxo Oncology, Hutchinson MediPharma, Ideaya, Amgen, Tango, Mirati, Linnaeus, Monte Rosa, Kinnate, Debio, BioTheryX, Storm, BeiGene, MapKure, Relay, Novartis, Fusion Pharma, C4, Scorpion, Incyte, Fog, Tyra, Nuvectis, BridgeBio, 3H Pharmaceuticals, AstraZeneca, Vall d'Hebron Institute of Oncology/Cancer Core Europe. **NAB**: Nothing to disclose.

RESULTS *(Cont'd)*

Treatment-Emergent Adverse Events in ≥20% Any Grade and/or ≥5% Grade ≥3 of Patients

	Preferred term	Overall (N=113) G1, n (%)	G2, n (%)	G3, n (%)	G4, n (%)	Any grade, n (%)	PCNST (n=22) G1, n (%)	G2, n (%)	G3, n (%)	G4, n (%)	Any grade, n (%)
Lab abnormality AEs	Increased ALT	24 (21.2)	14 (12.4)	9 (8.0)	1 (0.9)	48 (42.5)	9 (40.9)	2 (9.1)	4 (18.2)	0	15 (68.2)
	Increased AST	26 (23.0)	13 (11.5)	3 (2.7)	0	42 (37.2)	11 (50.0)	3 (13.6)	0	0	14 (63.6)
	Increased blood bilirubin	4 (3.5)	8 (7.1)	8 (7.1)	0	20 (17.7)	1 (4.5)	1 (4.5)	1 (4.5)	0	3 (13.6)
	Increased blood creatinine	14 (12.4)	4 (3.5)	0	0	18 (15.9)	8 (36.4)	0	0	0	8 (36.4)
	Hyponatremia	1 (0.9)	0	6 (5.3)	1 (0.9)	8 (7.1)	0	0	0	1 (4.5)	1 (4.5)
Symptomatic AEs	Fatigue	17 (15.0)	23 (20.4)	1 (0.9)	0	41 (36.3)	4 (18.2)	3 (13.6)	0	0	7 (31.8)
	Nausea	27 (23.9)	6 (5.3)	2 (1.8)	0	35 (31.0)	6 (27.3)	0	0	0	6 (27.3)
	Diarrhea	15 (13.3)	7 (6.2)	4 (3.5)	0	26 (23.0)	3 (13.6)	1 (4.5)	0	0	4 (18.2)
	Vomiting	11 (9.7)	11 (9.7)	1 (0.9)	0	23 (20.4)	1 (4.5)	3 (13.6)	0	0	4 (18.2)
	Headache	9 (8.0)	6 (5.3)	2 (1.8)	0	17 (15.0)	0	3 (13.6)	2 (9.1)	0	5 (22.7)

- The majority of symptomatic AEs were grade 1 in severity.
- Ocular toxicities or secondary skin cancers associated with early BRAFis[3] were not seen.
 - Eye-related TEAEs (ie, dry eye, blurred vision, pruritus) were reported in <5% of patients.
- Grade 4 events were very rare and no fatal treatment-related adverse events occurred.

RP2D

- Evaluated total daily doses 900–3600 mg with or without cobicistat 150 mg coadministered QD (PK enhancer that increased plixorafenib exposure 2- to 3-fold).
- Clinically relevant exposures were observed across dose levels with or without cobicistat, suggesting a broad therapeutic window.
- Based on the totality of the data for safety, PK, pharmacodynamics, and efficacy, the optimal RP2D was determined to be 900 mg QD coadministered with cobicistat for ages ≥10 years.

Efficacy in Patients With V600-Mutated Tumors

Excluding CRC and patients who received BSA-adjusted dosing

- ORR, n (%; 95% CI): 13 (30.2%; 17.2, 46.1); Median (range) DOR: 17.8 (3.7, 59.2) months
- Median (range) time to response: 3.6 (1.7, 81.2) months
- 1-year PFS rate: 32.6%; 2-year PFS rate: 16.3% (in the MAPKi-naïve subgroup: 1- and 2-year PFS were 45.8% and 29.2%, respectively)
- Antitumor activity was observed in various tumor types.

Plixorafenib Best Percent Tumor Change From Baseline in V600+ Adults (N=43, mITT)[a]

Excluding CRC due to known intrinsic resistance through EGFR pathway

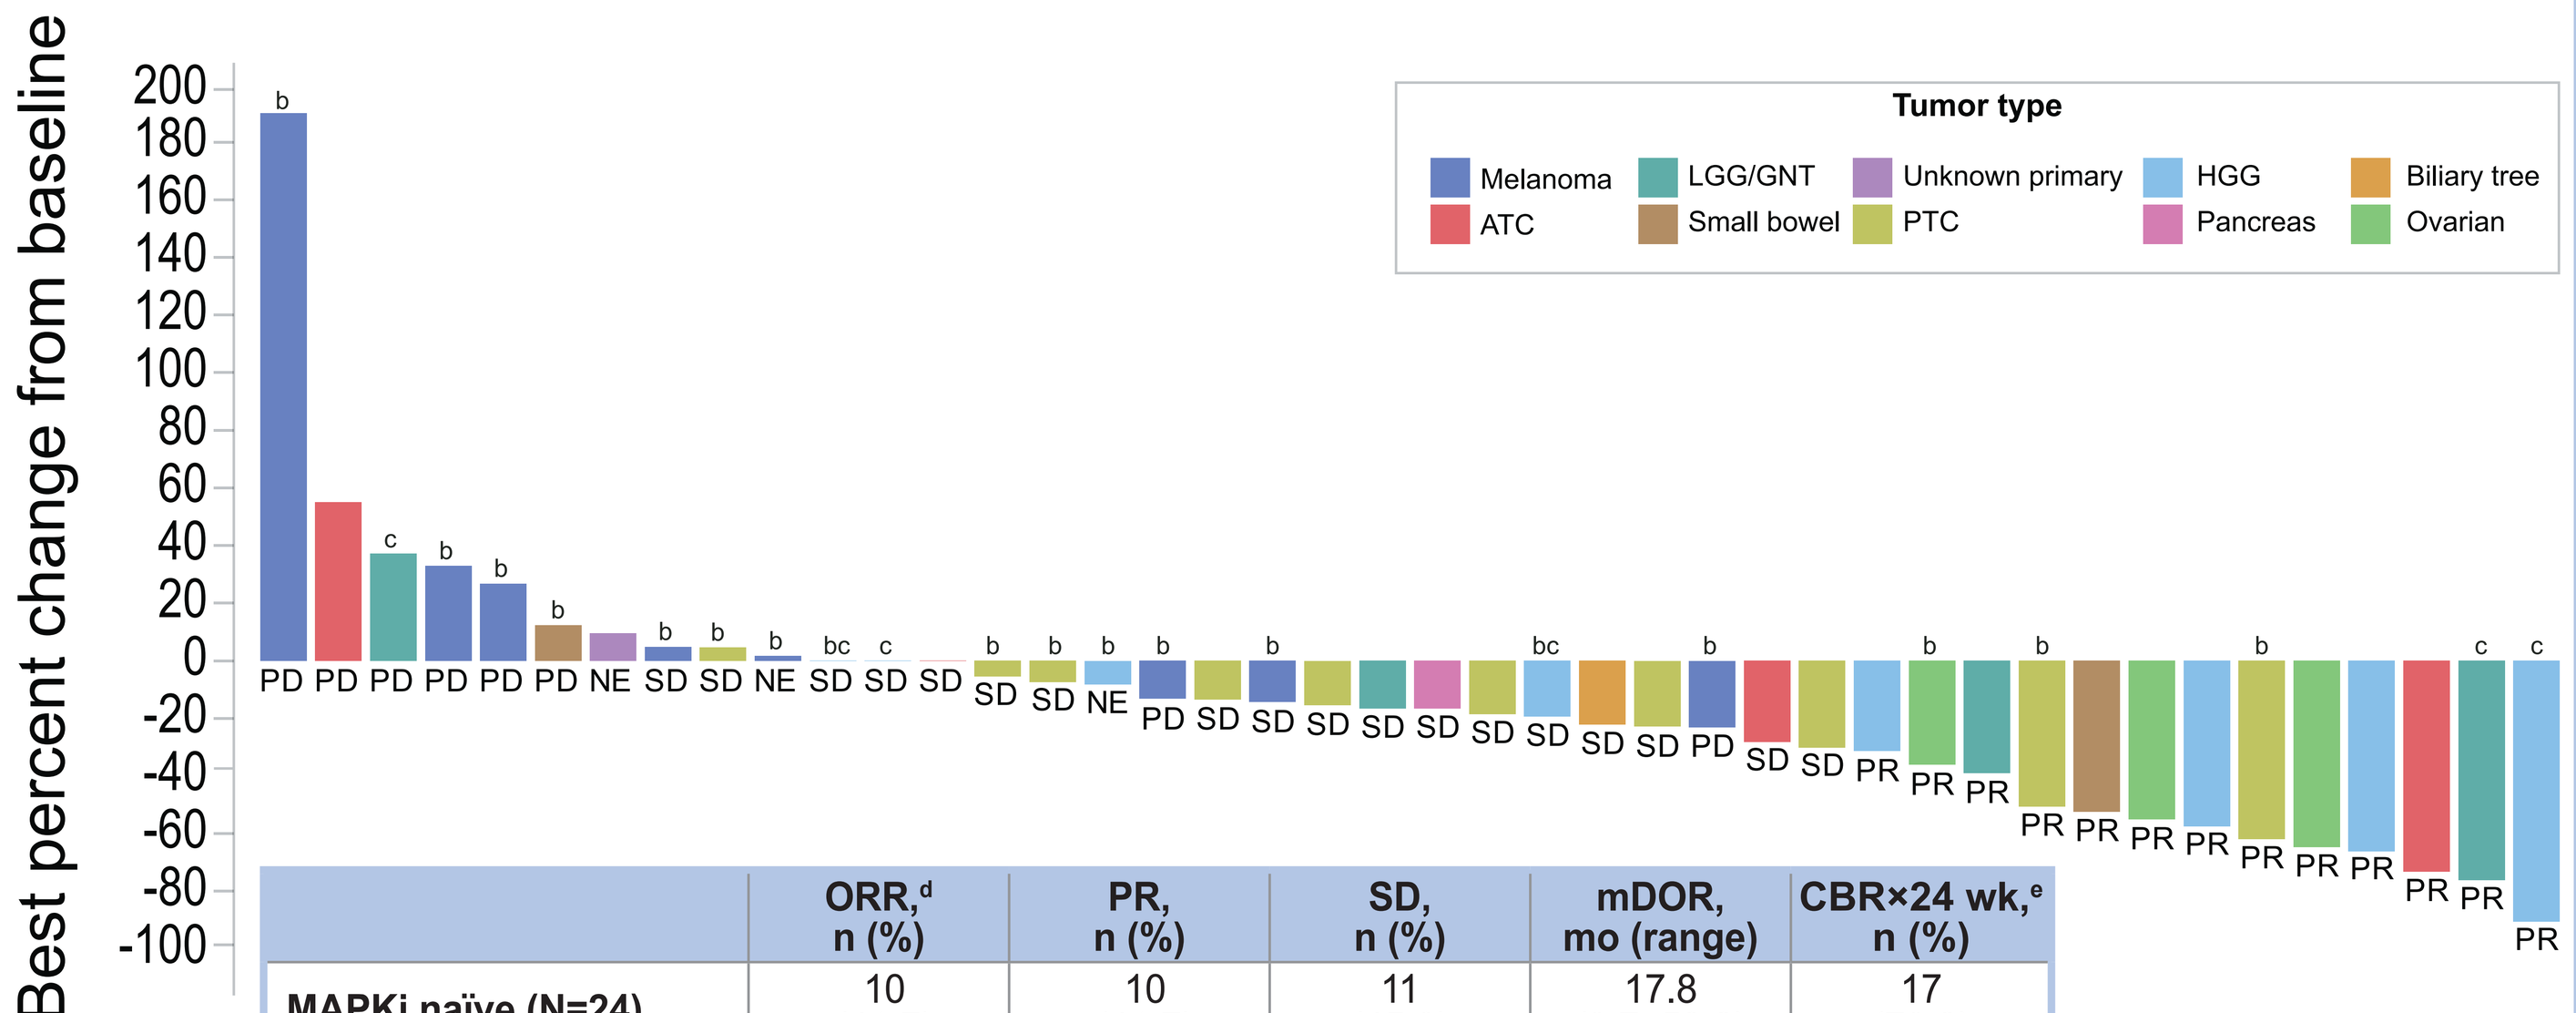
Best percent change from baseline
200
180
160
140
120
100
80
60
40
20
0
-20
-40
-60
-80
-100
Tumor type
Melanoma
ATC
LGG/GNT
Small bowel
Unknown primary
PTC
HGG
Pancreas
Biliary tree
Ovarian
PD PD PD PD PD PD NE SD SD NE SD SD SD SD SD NE PD SD SD SD SD SD SD SD SD SD PD SD SD PR PR PR PR PR PR PR PR PR PR PR PR PR PR

	ORR,[d] n (%)	PR, n (%)	SD, n (%)	mDOR, mo (range)	CBR×24 wk,[e] n (%)
MAPKi naïve (N=24)	10 (41.7)	10 (41.7)	11 (45.8)	17.8 (3.7, 59.2)	17 (70.8)
MAPKi pretreated (N=19)	3 (15.8)	3 (15.8)	7 (36.8)	12.9 (3.9, 16.6+)	7 (36.8)

[a]One patient was excluded due to the unavailability of postbaseline assessment target lesion measurement and best response was not evaluable.
[b]Indicates prior MAPKi treatment.
[c]Reported per RANO criteria.
[d]All responses were confirmed.
[e]CBR: CR+PR+MR+(SD≥24 weeks).

Plixorafenib Treatment Duration

Efficacy-evaluable adults with V600+ tumors

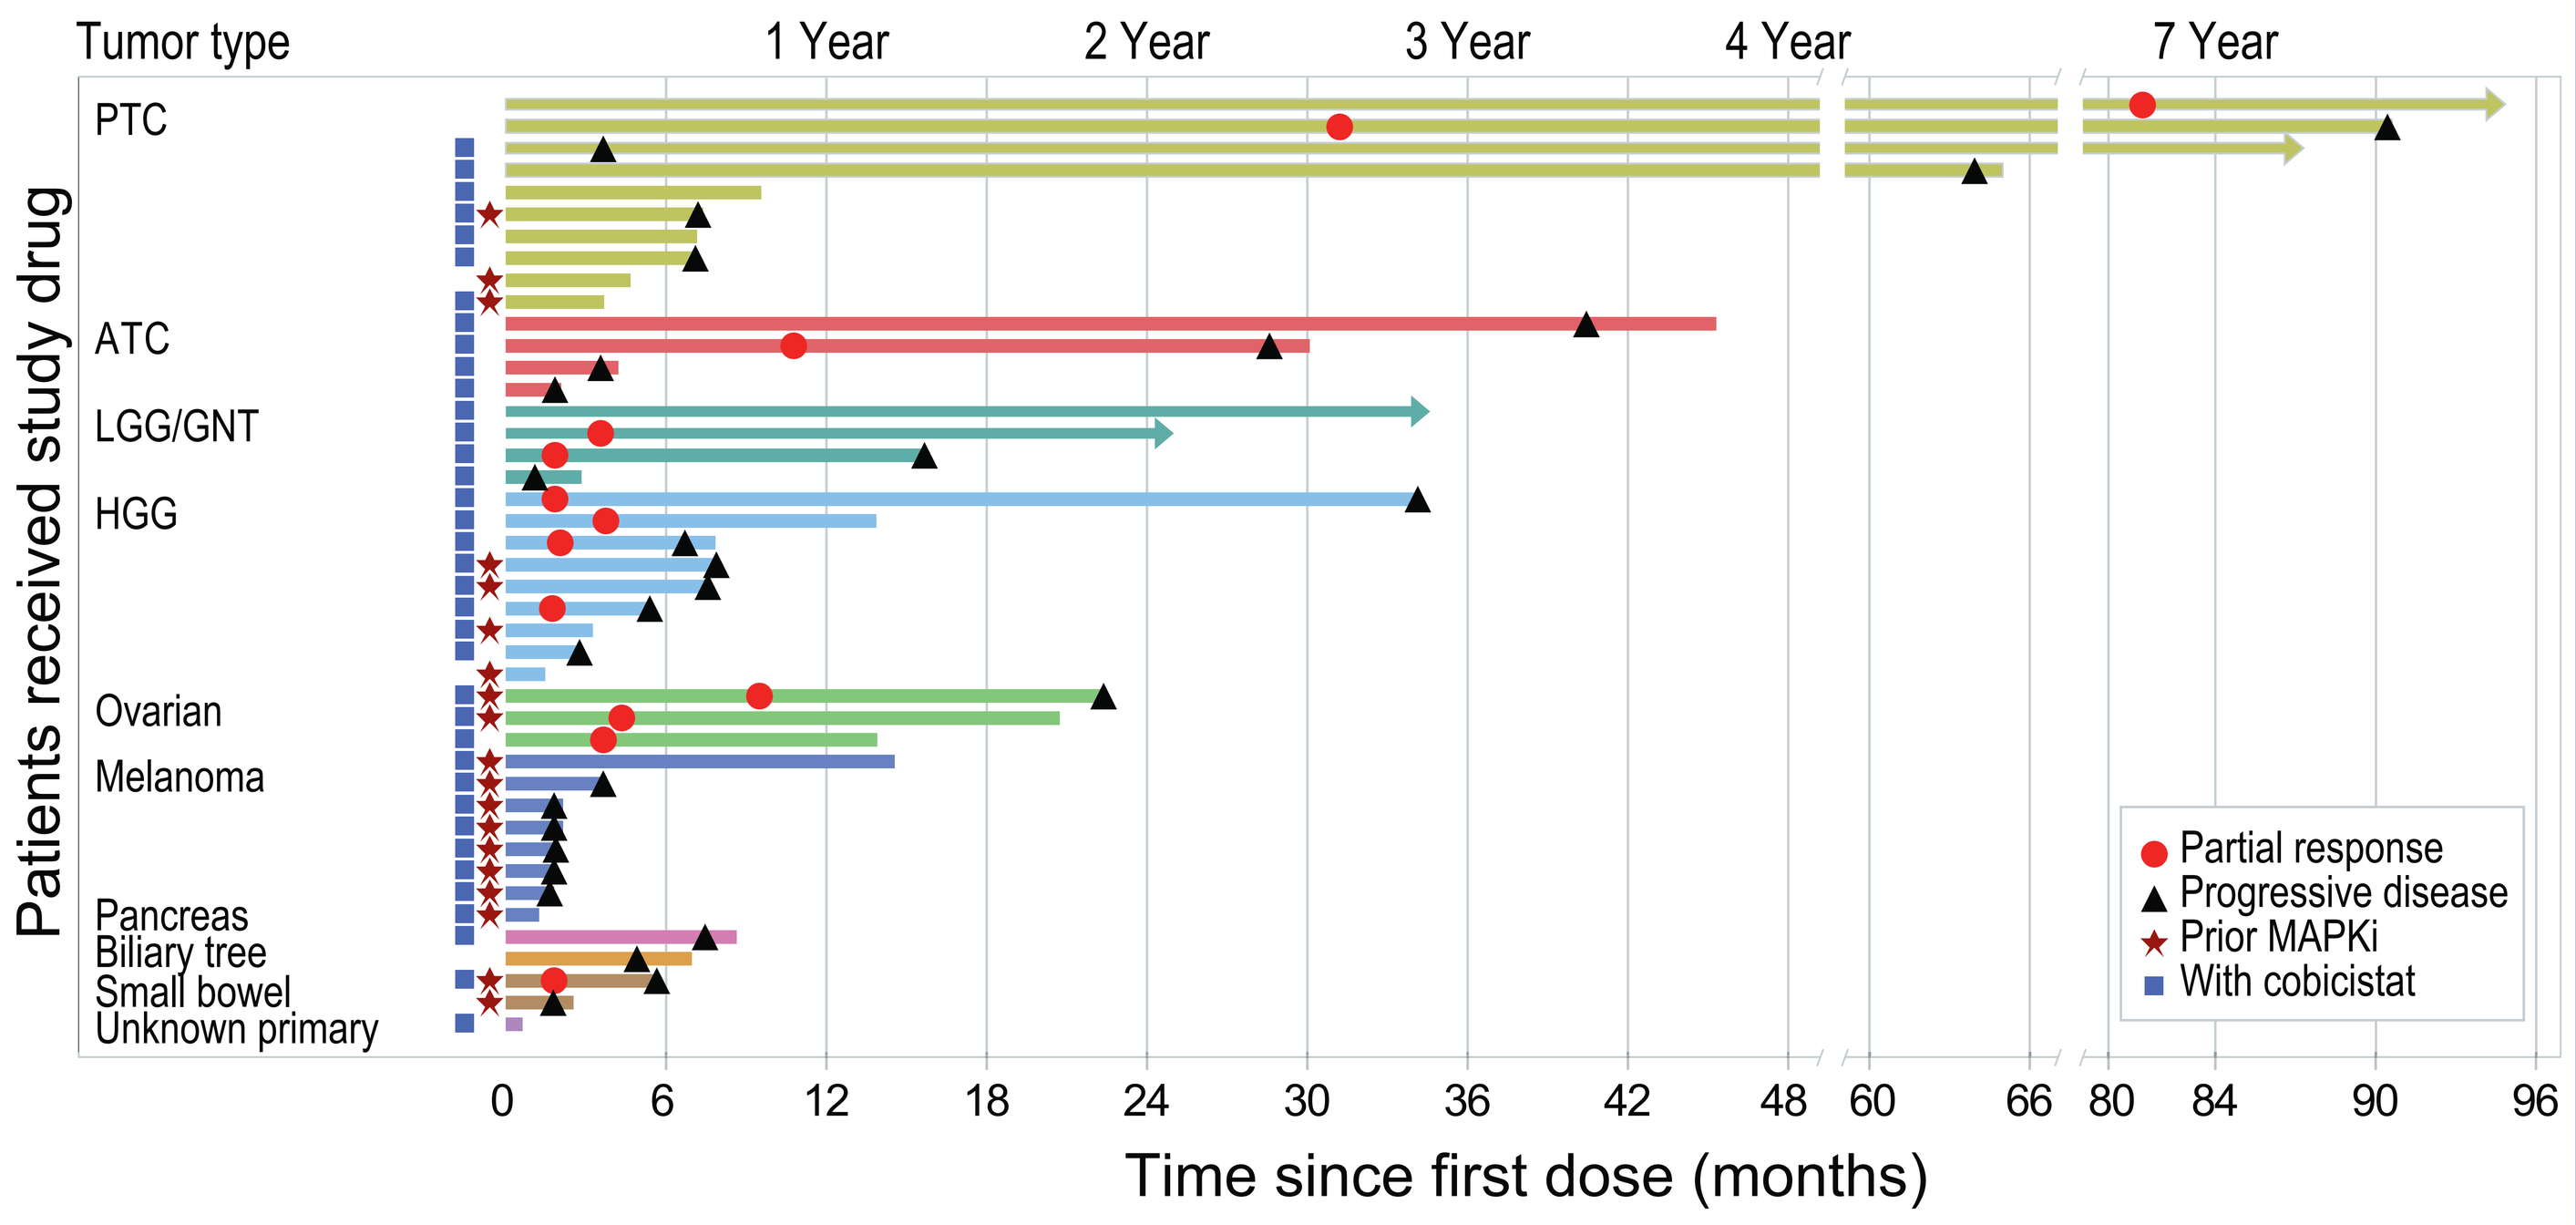
Tumor type
1 Year
2 Year
3 Year
4 Year
7 Year
PTC
ATC
LGG/GNT
HGG
Ovarian
Melanoma
Pancreas
Biliary tree
Small bowel
Unknown primary
Patients received study drug
Partial response
Progressive disease
Prior MAPKi
With cobicistat
0 6 12 18 24 30 36 42 48 60 66 80 84 90 96
Time since first dose (months)

Efficacy in BRAF Fusion Tumors

- ORR: 14.3% (2/14) of adults
 - 1 with melanoma: CR, DOR 66.7+ months
 - 1 with PTC: PR, DOR 9.2+ months
 - Of the 8 with SD, 5 had PCNST
 - 1 with GBM and SD remained on treatment for 7.5 months
 - 5 had PD (3 with co-occurring driver mutations)

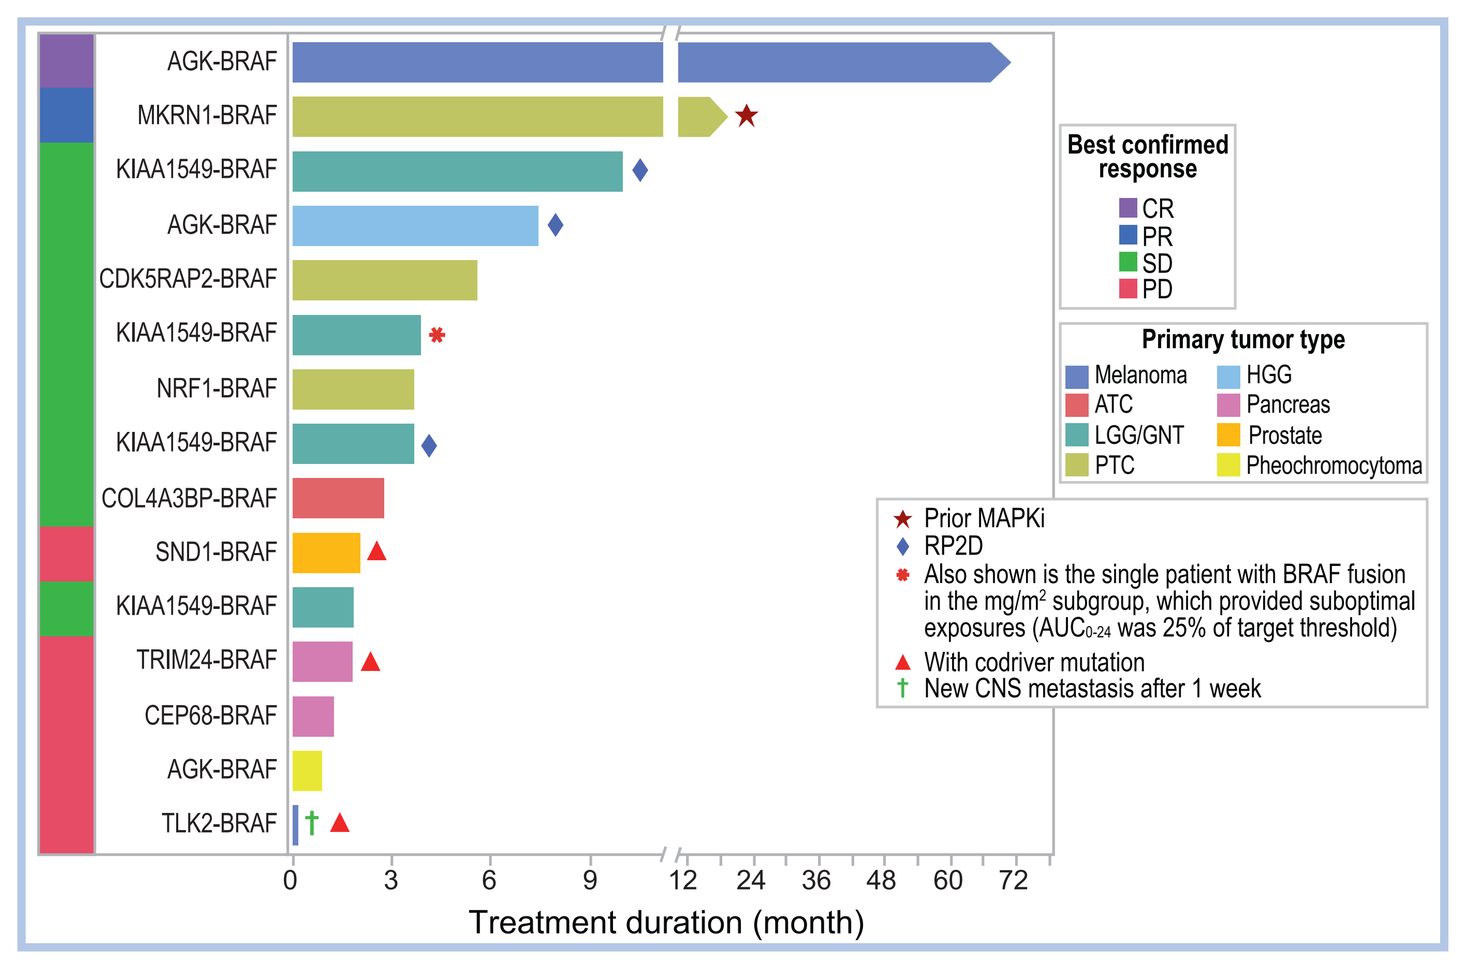
AGK-BRAF
MKRN1-BRAF
KIAA1549-BRAF
AGK-BRAF
CDK5RAP2-BRAF
KIAA1549-BRAF
NRF1-BRAF
KIAA1549-BRAF
COL4A3BP-BRAF
SND1-BRAF
KIAA1549-BRAF
TRIM24-BRAF
CEP68-BRAF
AGK-BRAF
TLK2-BRAF
Best confirmed response
CR
PR
SD
PD
Primary tumor type
Melanoma
ATC
LGG/GNT
PTC
HGG
Pancreas
Prostate
Pheochromocytoma
Prior MAPKi
RP2D
Also shown is the single patient with BRAF fusion in the mg/m² subgroup, which provided suboptimal exposures (AUC0-24 was 25% of target threshold)
With codriver mutation
New CNS metastasis after 1 week
0 3 6 9 12 24 36 48 60 72
Treatment duration (month)

Pediatric Demographics, Clinical Characteristics, and Treatment History

Age range, year	Sex, n (%) Female	Male	Tumor types	Prior lines of treatment, median (range)	Plixorafenib BID dosing Range, mg/m²	Flat, mg
4–17	2 (40)	3 (60)	• LGG (n=3)[ab] • Neuroblastoma (n=1) • LCH-ND (n=1)[c]	5 (4–8)	250–500	600

[a]Two patients with pilocytic astrocytoma and 1 with pilomyxoid astrocytoma.
[b]One patient (LGG; pilocytic astrocytoma) had an amplification and was excluded from the efficacy-evaluable population.
[c]After 40 weeks of treatment, the dose for the patient with LCH-ND was increased from 250 mg/m² to 375 mg/m² BID.

Safety and Efficacy in Pediatric Patients

- No DLTs, dose reductions, or LFT elevations occurred.
- One patient with LGG (pilocytic astrocytoma) reported a grade 3 headache that was possibly related to plixorafenib; no other ≥ grade 3 AEs related to plixorafenib were reported.
- Three patients (2 with BRAF V600, 1 with BRAF fusion) receiving BSA-adjusted dosing had SD; 1 patient with LCH-ND has been on treatment for 72.6+ months.
- The fourth evaluable patient with neuroblastoma and Class II BRAF mutation (nonfusion) who received 600 mg BID had PD.

Clinical Pharmacokinetics

- Dose-adjusted AUC and C_{max} were comparable between adult and pediatric patients.
- PK exposure was not dependent on age (4–86 years) or weight (18–132 kg) across patients.

Simulated Steady-state AUC_{0-24} by Weight[a]

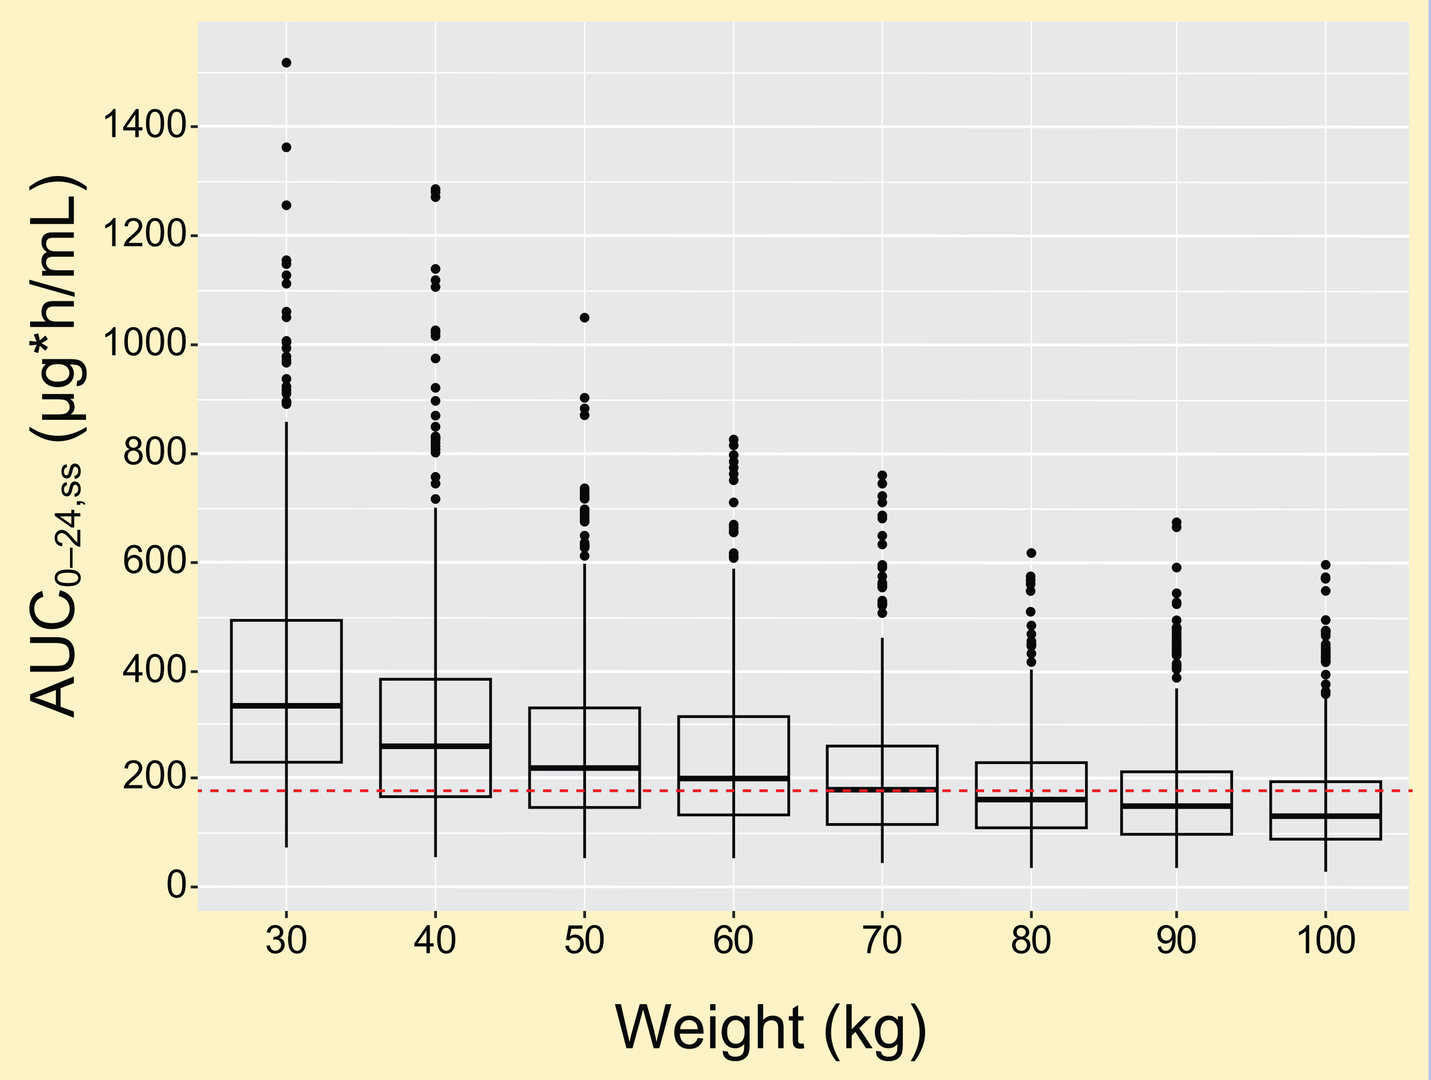
AUC0–24,ss (μg*h/mL)
1400
1200
1000
800
600
400
200
0
30 40 50 60 70 80 90 100
Weight (kg)

[a]RP2D was used for modeling.

Efficacy in V600+ MAPKi-naïve PCNST

- ORR: 6/9 had PR (66.7%; 95% CI: 29.9–92.5)
- CBR (response or SD×24 weeks): 78%
- Median (range) duration of response: 13.9 (3.7, 32.3) months
- Response in patients with only nonenhancing lesions and/or pilocytic astrocytoma demonstrate that clinically relevant CNS penetration is achieved.

	LGG (n=4)	HGG (n=5)
Confirmed response, n (%)		
PR	2 (50.0)	4 (80.0)
SD	1 (25.0)	1 (20.0)
PD	1 (25.0)	0
ORR, n (%)	2 (50.0)	4 (80.0)
DOR range, mo	13.9, 16.6+	3.7, 32.3
6-mo DOR rate, n (%)[a]	2/2 (100.0)	2/4 (50.0)
CBR×24 wk[b]	3 (75.0)	4 (80.0)
mPFS, mo (range)	NE (1.6, 33.6+)	6.7 (2.8, 34.1)

[a]≥6-month DOR rate: % of responders with DOR ≥6 months.
[b]CBR×24 wk: CR+PR+MR+(SD≥24 weeks).

Efficacy and Treatment Duration by Patient

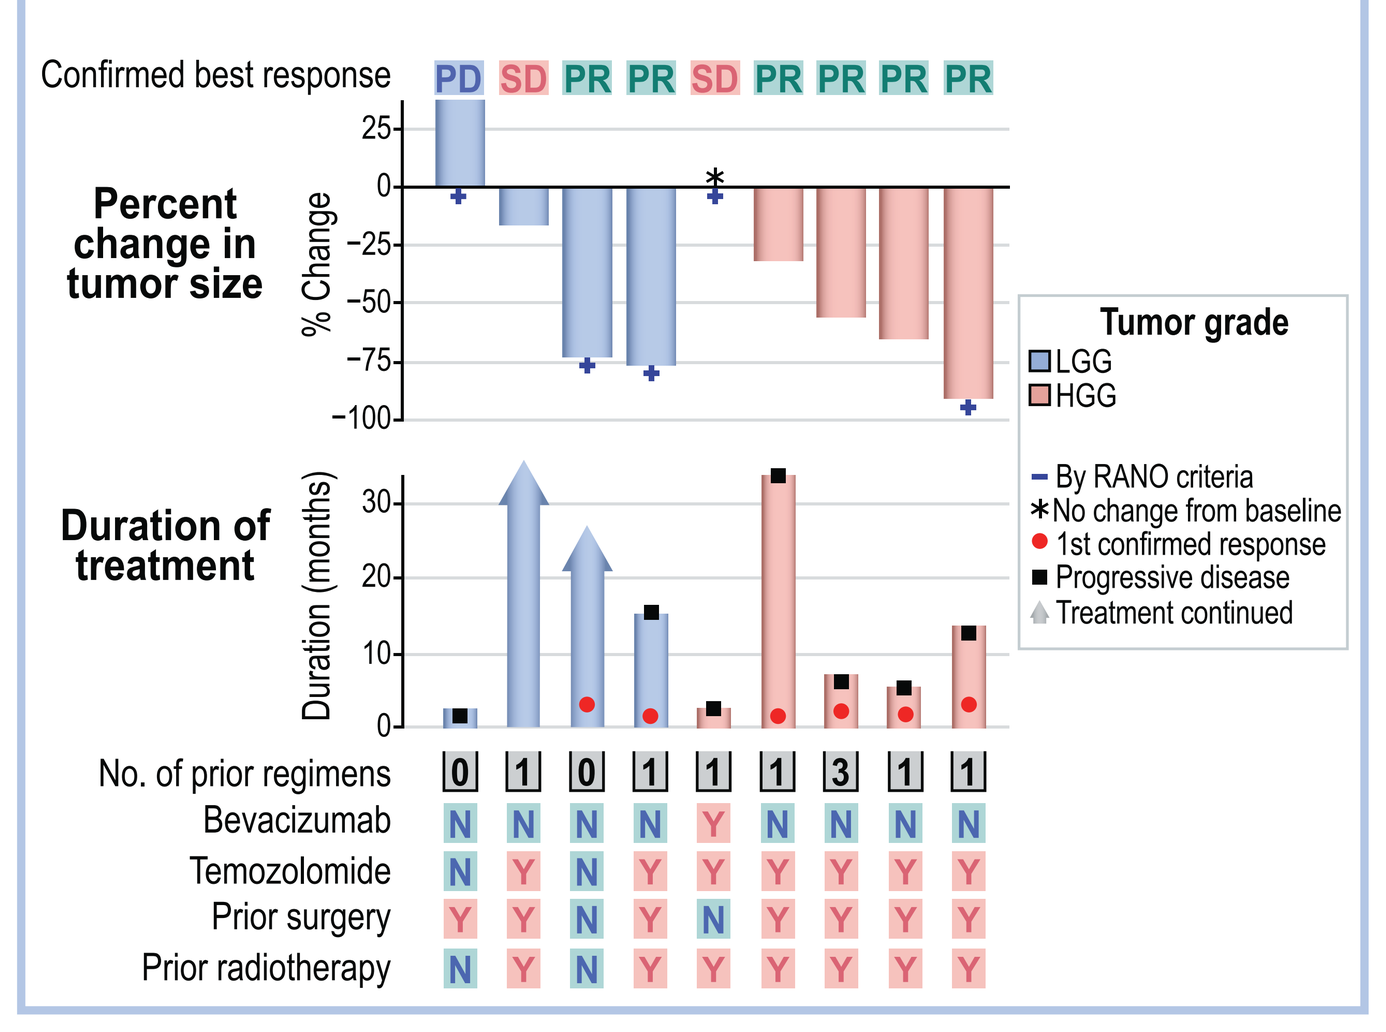
Confirmed best response
PD SD PR PR SD PR PR PR PR
Percent change in tumor size
% Change
Tumor grade
LGG
HGG
By RANO criteria
No change from baseline
1st confirmed response
Progressive disease
Treatment continued
Duration of treatment
Duration (months)
No. of prior regimens 0 1 0 1 1 1 3 1 1
Bevacizumab N N N N Y N N N N
Temozolomide N Y N Y Y Y Y Y Y
Prior surgery Y Y N Y N Y Y Y Y
Prior radiotherapy N Y N Y Y Y Y Y Y

Genomic Data in PCNST

- Baseline genomic data was collected for 13 adults (MAPKi naïve, n=9; MAPKi pretreated, n=4).
- PR observed in patients with:
 - CDKN2A/B deletion (n=4)
 - PTEN deletion (n=1)
 - MTAP deletion (n=2)
- An increase in molecular heterogeneity was not observed between MAPKi-naïve- and MAPKi-pretreated subgroups.

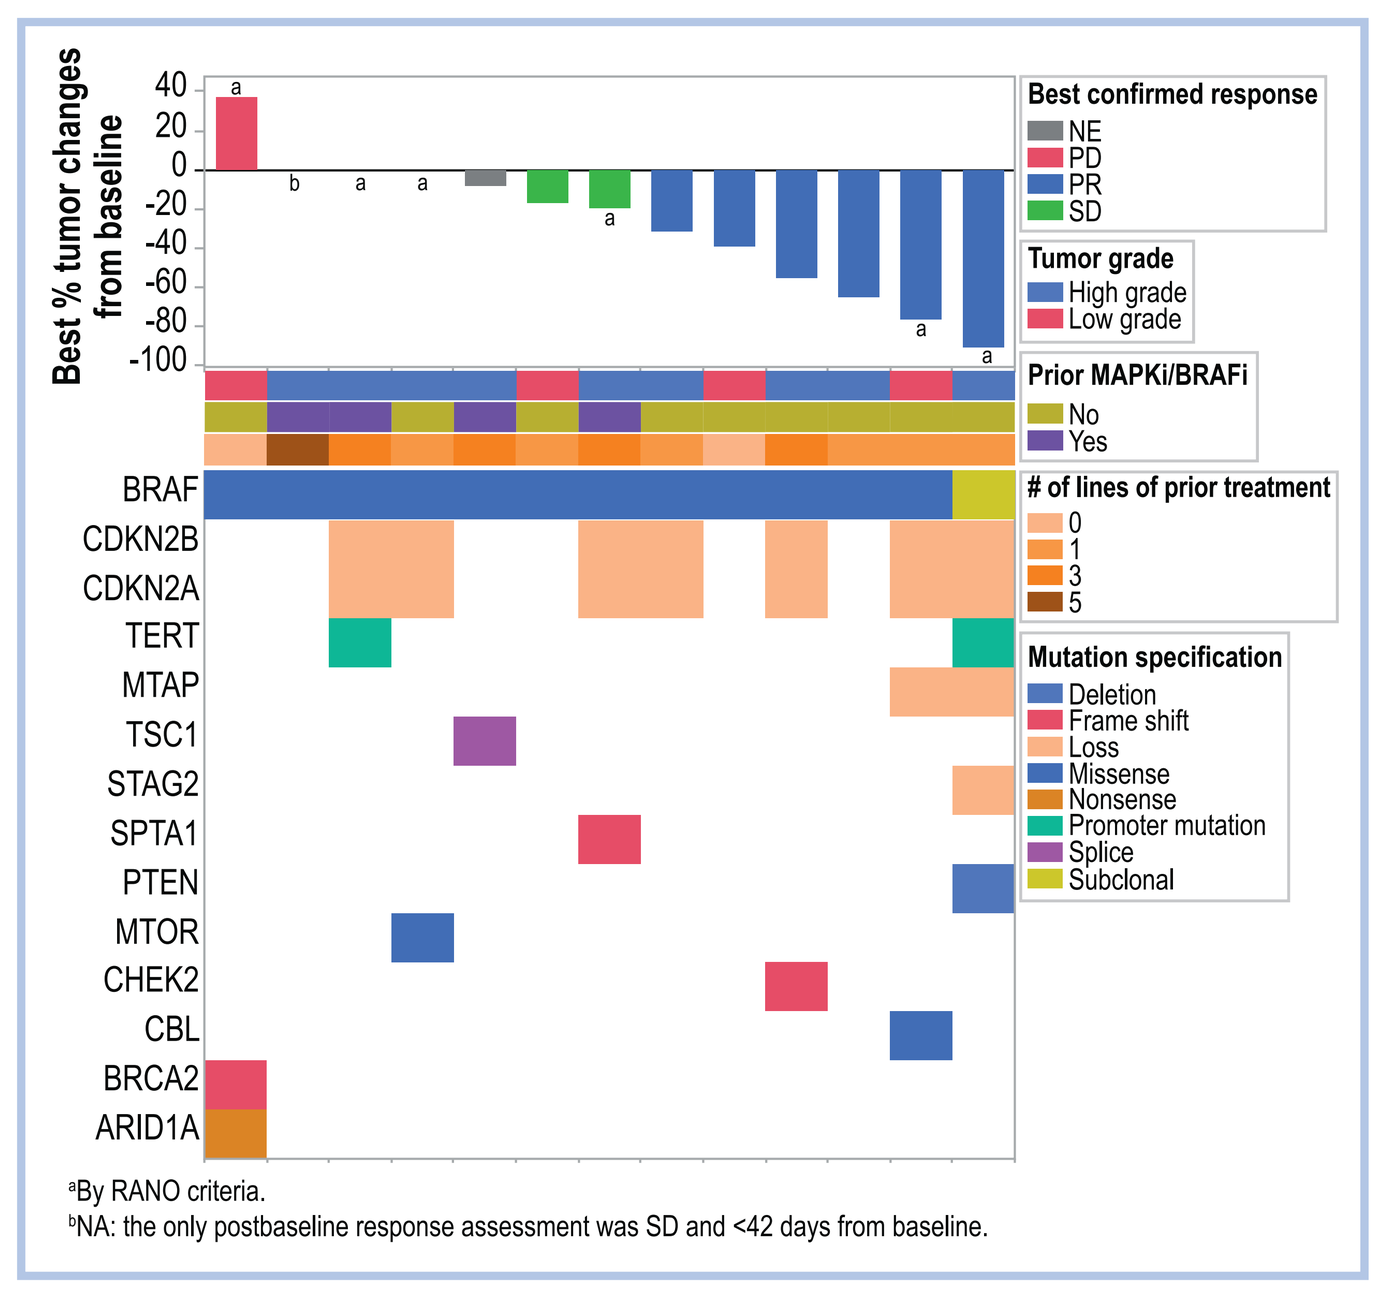
Best % tumor changes from baseline
Best confirmed response
NE
PD
PR
SD
Tumor grade
High grade
Low grade
Prior MAPKi/BRAFi
No
Yes
of lines of prior treatment
0
1
3
5
Mutation specification
Deletion
Frame shift
Loss
Missense
Nonsense
Promoter mutation
Splice
Subclonal
BRAF
CDKN2B
CDKN2A
TERT
MTAP
TSC1
STAG2
SPTA1
PTEN
MTOR
CHEK2
CBL
BRCA2
ARID1A

[a]By RANO criteria.
[b]NA: the only postbaseline response assessment was SD and <42 days from baseline.

CONCLUSIONS

- Plixorafenib was well tolerated with low rates of symptomatic TEAEs.
- In the 5 pediatric patients, there were minimal related AEs with the overall population, no DLTs, and no LFT changes.
- Antitumor activity was observed across populations, with 6/9 PRs (67% ORR) in patients with MAPKi-naïve BRAF V600 PCNST.
- A phase 2 study of children and adults with BRAF V600E-mutated PCNST is ongoing (NCT05503797).

Abbreviations

AE, adverse event; ALT, alanine transaminase; AST, aspartate transaminase; ATC, anaplastic thyroid cancer; AUC, area under the curve; BRAF, v-raf murine sarcoma viral oncogene homolog B1; BRAFi, BRAF inhibitor; BSA, body surface area; CBR, clinical benefit rate; CI, confidence interval; C_{max}, maximum serum concentration; CNS, central nervous system; CR, complete response; CRC, colorectal cancer; DLT, dose-limiting toxicity; DOR, duration of response; EGFR, epidermal growth factor receptor; GBM, glioblastoma; GNT, glioneuronal tumor; HGG, high-grade glioma; LFT, liver function test; LGG, low-grade glioma; MAPK, mitogen-activated protein kinase; MEK, MAPK kinase; mDOR, median DOR; mITT, modified intent to treat; mPFS, median PFS; MR, minor response; NA, not applicable; LCH-ND, neurodegenerative Langerhans cell histiocytosis; NE, not evaluable; NSCLC, non–small cell lung cancer; ORR, overall response rate; PCNST, primary central nervous system tumor; PD, progressive disease; PFS, progression-free survival; PK, pharmacokinetic; PR, partial response; PTC, papillary thyroid carcinoma; QD, once daily; RP2D, recommended phase 2 dose; SD, stable disease.

References

1. Yao Z et al. *Nat Med* 2019;25:284–91; 2. Tutuka CSA et al. *Mol Cancer* 2017;16:112; 3. Welsh SJ and Corrie PG. *Ther Adv Med Oncol* 2015;7:122–36; 4. Hanrahan AJ et al. *Nat Rev Clin Oncol* 2024;21:224–47; 5. Ostrom QT et al. *Neuro Oncol* 2023;25(S4):iv1–iv99.

Acknowledgments

Funding for this study (NCT02428712 available from www.clinicaltrials.gov) was provided by Fore Biotherapeutics (Fore Bio). Medical editorial support (Lacy Miron, PharmD) and graphic services were provided by AOIC, LLC and were funded by Fore Bio.

Presented at the International Symposium on Pediatric Neuro-Oncology (ISPNO 2024) • Philadelphia, PA, USA • June 28–July 2, 2024